DEVELOPMENT OF NOVEL, SELECTIVE SMARCA2 (BRM) DEGRADERS FOR TREATMENT OF SMARCA4 (BRG1) MUTATED TUMORS

RYVU THERAPEUTICS

AACR American Association for Cancer Research® VIRTUAL ANNUAL MEETING II JUNE 22-24, 2020

Andrzej Mazan, Anna Wróbel, Agnieszka Dreas, Adam Radzimierski, Kinga Michalik, Anna Kowal, Katarzyna Wiklik, Katarzyna Wnuk-Lipińska, Paulina Niedziejko, Kamila Kozłowska, Magdalena Łośko, Joanna Szczęśniak, Agnieszka Sroka-Porada, Justyna Martyka, Urszula Kulesza, Karol Zuchowicz, Magdalena Zastawna, Jose Alvarez, Luigi Stasi, Peter Littlewood,Tomasz Rzymski, Krzysztof Brzózka.

Contact: andrzej.mazan@ryvu.com; Ryvu Therapeutics S.A. Bobrzyńskiego 14, 30-348 Kraków, Poland

Poster Number: #3656

SUMMARY

Synthetic lethality is one of the most innovative approaches for selective targeting of cancer cells with defined genetic background. SMARCA2 and SMARCA4 are two mutually exclusive helicase/ATPase catalytic subunits belonging to SWI/SNF chromatin remodeling complex. It is estimated that one in five solid tumors carries a mutation in proteins of this complex. The Cancer Genome Atlas (TCGA) and the Catalog of Somatic Mutation in Cancer (Cosmic) show that SMARCA4 is one of the most mutated genes in lung, colorectal, breast, melanoma and CNS tumors. Herein, we present development of a potent and selective SMARCA2 degrader, structurally unrelated to known chemotypes with first in class potential selectively targeting SMARCA4 mutant cells.

INTRODUCTION

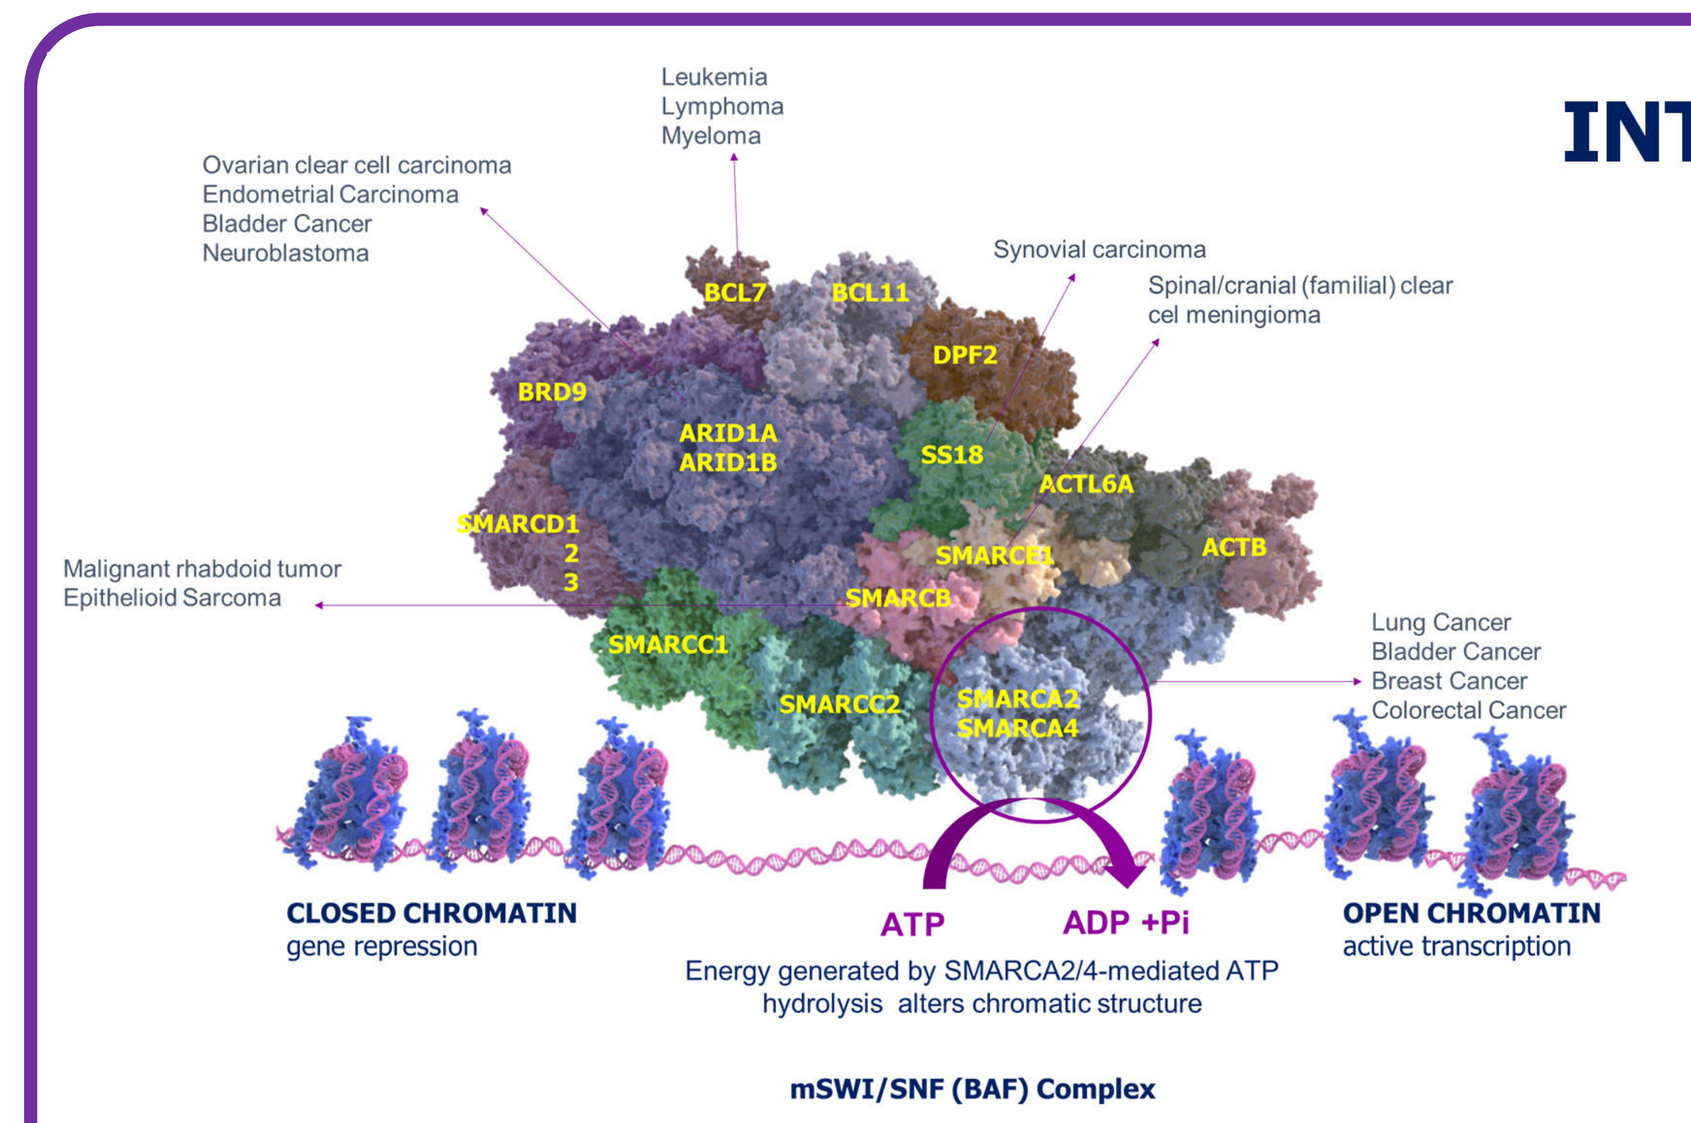

Leukemia
Lymphoma
Myeloma
Ovarian clear cell carcinoma
Endometrial Carcinoma
Bladder Cancer
Neuroblastoma
Synovial carcinoma
Spinal/cranial (familial) clear cel meningioma
Malignant rhabdoid tumor
Epithelioid Sarcoma
Lung Cancer
Bladder Cancer
Breast Cancer
Colorectal Cancer
CLOSED CHROMATIN
gene repression
ATP
ADP +Pi
OPEN CHROMATIN
active transcription
Energy generated by SMARCA2/4-mediated ATP hydrolysis alters chromatic structure
mSWI/SNF (BAF) Complex

Figure 1. SMARCA2 and SMARCA4 are catalytic subunits of SWI/SNF complex frequently mutated in cancers. Adapted from: Kadoch, C., Copeland, R. A., & Keilhack, H. (2016) *Biochemistry*, *55* (11), 1600-1614

SMARCA2/4, core members of SWI/SNF complex, are two mutually exclusive ATP-dependent engines that function in mobilizing nucleosomes to regulate transcription, DNA replication and repair. While SMARCA4 KO (knock-out) mice are embryonic lethal and conditional KO mice exhibit heart complications and GI track defects, SMARCA2 KO are viable with no obvious phenotype.

SMARCA4 is one of the most often mutated gene in NSCLC (TCGA). Genetic silencing studies demonstrated that SMARCA4 LOF cells are addicted to SMARCA2 expression. Thus, development of SMARCA2 selective inhibitors/degraders is an attractive approach for cancer therapy with unmet medical need either as single agent or in combination with radio- or chemotherapies.

Here we present development and characterization of selective SMARCA2 degrader inducing apoptosis and growth inhibition predominantly in SMARCA4 loss-of-function (LOF) cells in proteasome dependent manner.

SMARCA4 LOF TUMORS ARE ADDICTED TO SMARCA2 EXPRESSION

a) Bioinformatic analysis

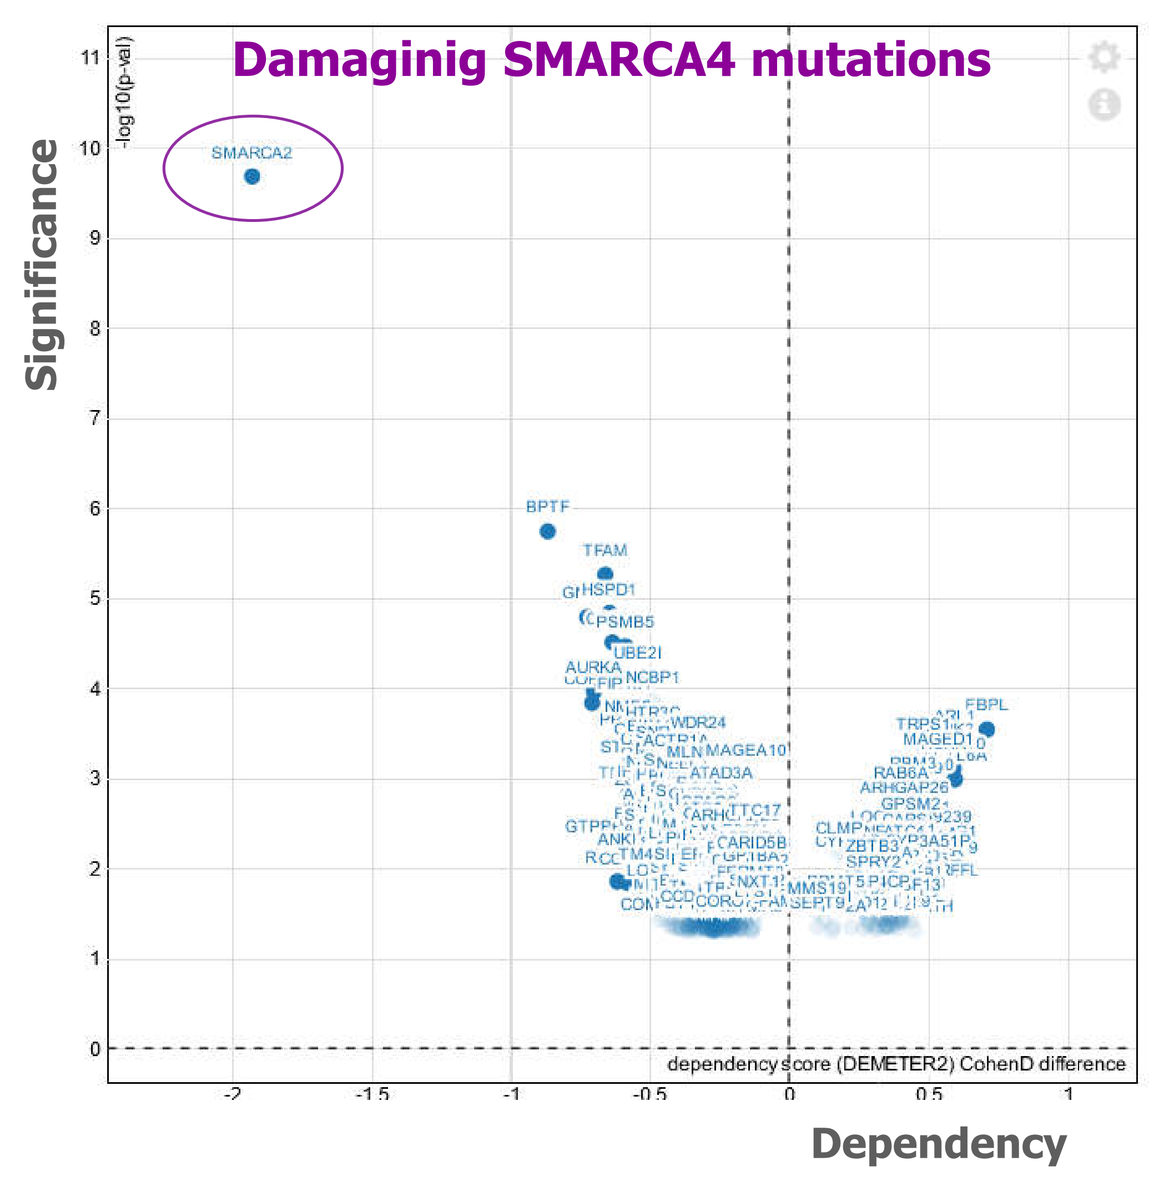

Damaginig SMARCA4 mutations
SMARCA2
Significance
Dependency

b) Frequency of cell lines sensitive to SMARCA2 silencing

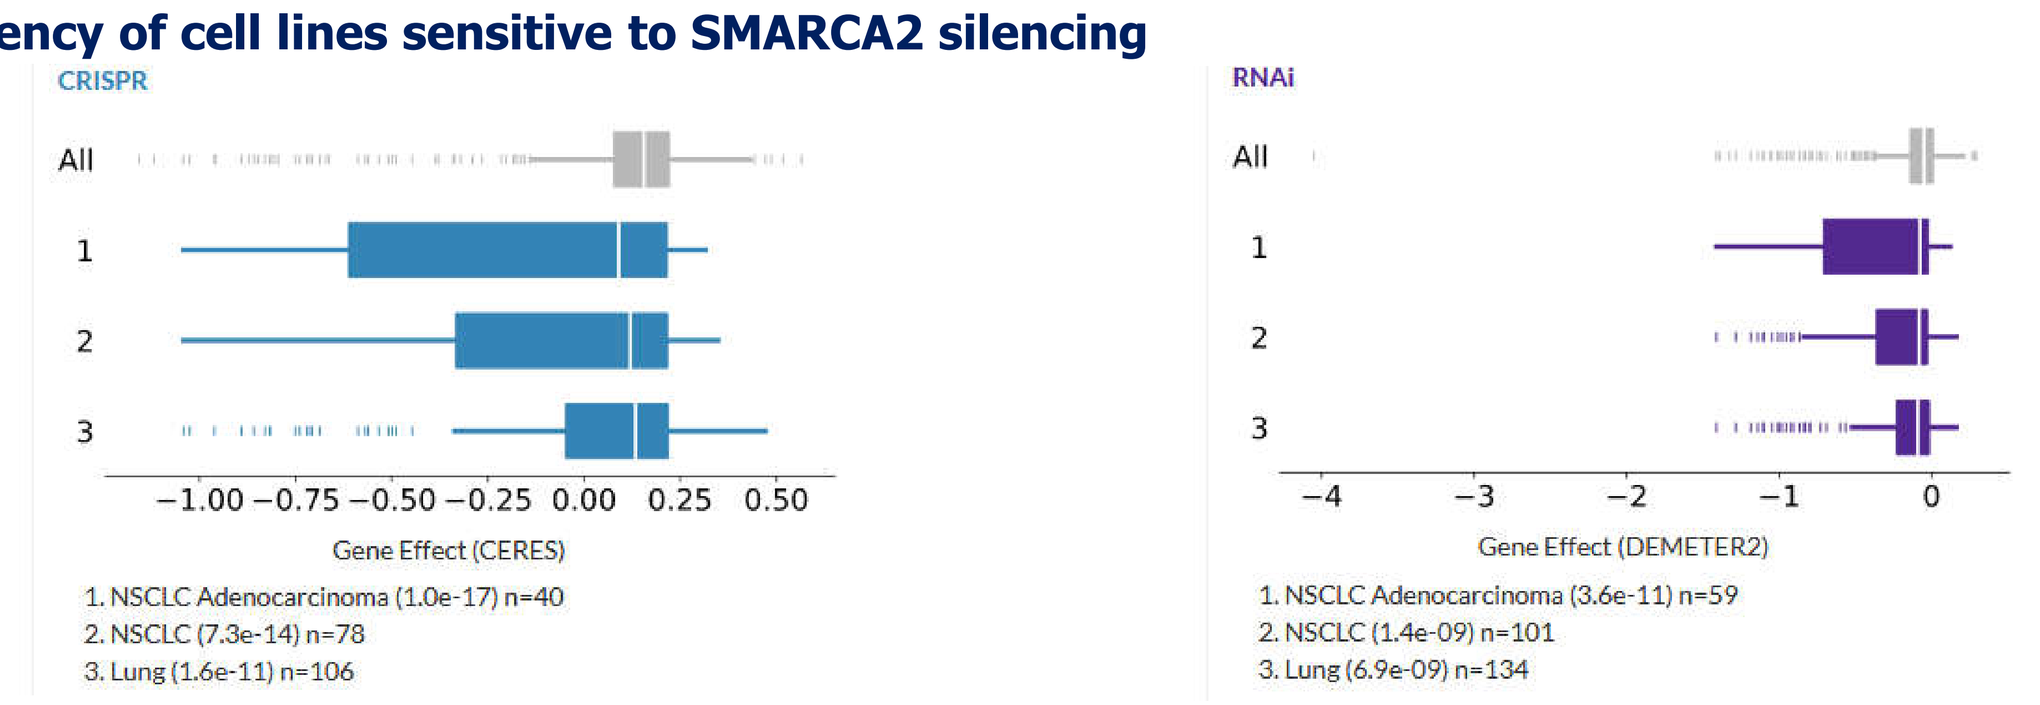

CRISPR
Gene Effect (CERES)
1. NSCLC Adenocarcinoma (1.0e-17) n=40
2. NSCLC (7.3e-14) n=78
3. Lung (1.6e-11) n=106
RNAi
Gene Effect (DEMETER2)
1. NSCLC Adenocarcinoma (3.6e-11) n=59
2. NSCLC (1.4e-09) n=101
3. Lung (6.9e-09) n=134

c) Experimantal validation of SMARCA4 WT and KO isogenic pair

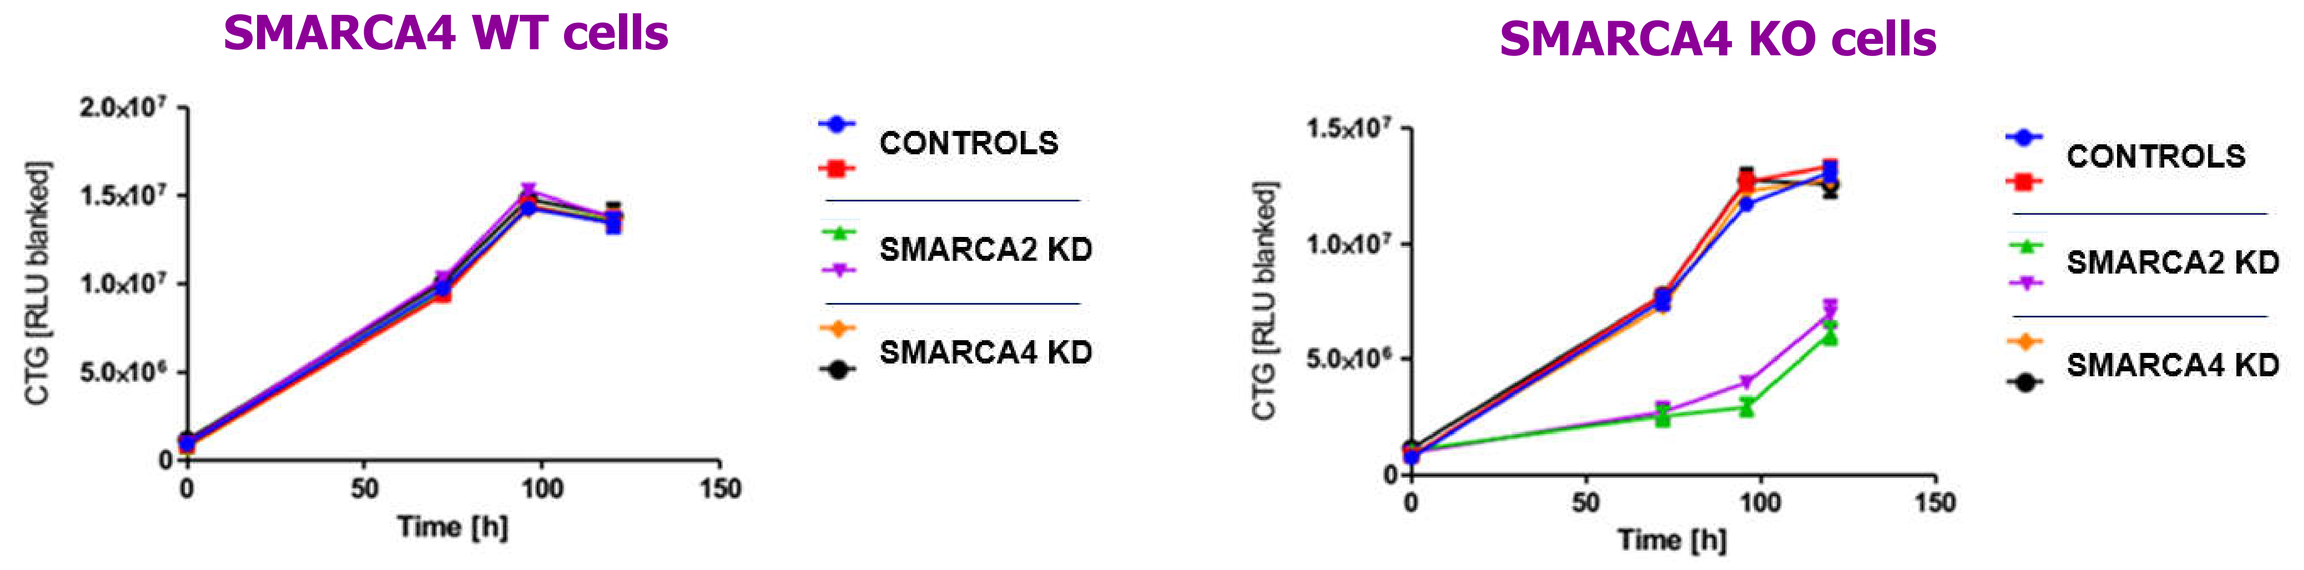

SMARCA4 WT cells
SMARCA4 KO cells
CONTROLS
SMARCA2 KD
SMARCA4 KD
CTG [RLU blanked]
Time [h]

Figure 2a. Ryvu proprietary bioinformatic tool: MultiDep identified SMARCA2 as a unique dependency in SMARCA4 cells carrying damaging mutations in SMARCA4 gene.

Figure 2b. Pan-cancer analysis showing enriched lineages sensitive to RNAi or CRISPR mediated SMARCA2 inhibition. Enriched lineages have p-values < 0.0005 (shown in parentheses). n= indicates the number of cell lines plotted in that lineage (source: DepMap).

Figure 2c. Homozygous LOF mutation was introduced in HT1080 WT cells uisng CRISPR/Cas9 technology. siRNA mediated SMARCA2 or SMARCA4 did not affect viability of WT cells. However, SMARCA2 but not SMARCA4 knock-down significantly affected viability of SMARCA4 KO cells (values are expressed as means +/-SD).

IDENTIFICATION OF NOVEL ALLOSTERIC INHIBITORS OF SMARCA2 ATPase ACTIVITY

a) Idetification of a hit compound

RVU	1771
hSMARCA2 pIC50	5.60
hSMARCA4 pIC50	4.90
MLM, Cl [ul/min/mg]	N.D.
HLM, Cl [ul/min/mg]	166.00
FTS Mean ΔTm @ 25uM	0.84
MST Kd [uM]	Binding confirmed
SPR Steady-state affinity analysis Kd	70 uM

RVU	2271
hSMARCA2 pIC50	6.5
hSMARCA4 pIC50	6.5
MLM, Cl [ul/min/mg]	357.0
HLM, Cl [ul/min/mg]	708.0
MST Kd [uM]	1.7

b) Medchem optimisation

RVU	2816
hSMARCA2 pIC50	6.6
hSMARCA4 pIC50	6.6
MLM, Cl [ul/min/mg]	56.0
HLM, Cl [ul/min/mg]	54.0
MST Kd [uM]	13.9

RVU	3626
hSMARCA2 pIC50	7.5
hSMARCA4 pIC50	7.4
MLM, Cl [ul/min/mg]	53.0
HLM, Cl [ul/min/mg]	62.0
MST DNA Kd [uM]	0.2

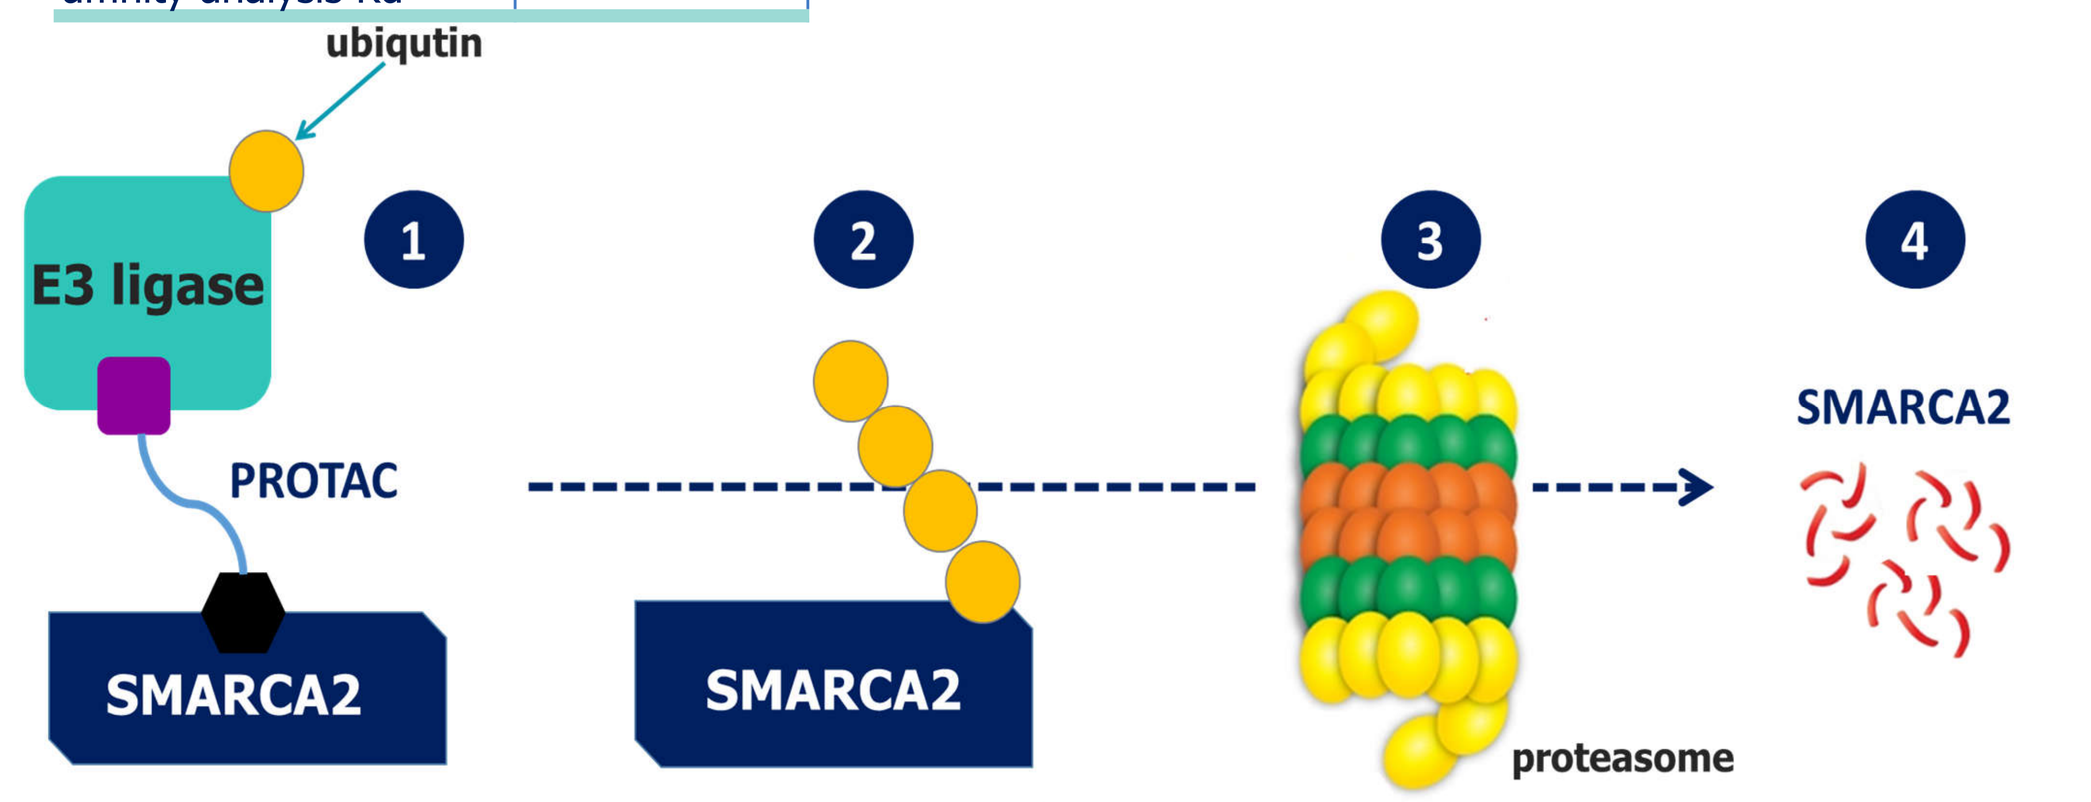

ubiqutin
E3 ligase
PROTAC
SMARCA2
1
2
3
4
SMARCA2
proteasome

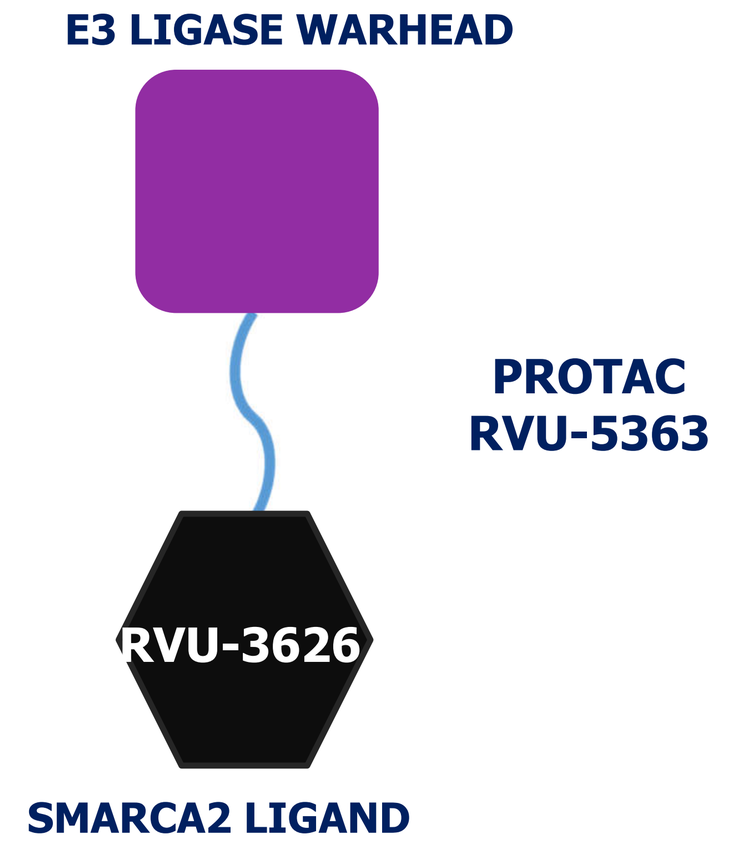

E3 LIGASE WARHEAD
PROTAC RVU-5363
RVU-3626
SMARCA2 LIGAND

Figure 3a. Biochemical HTS using SMARCA2 FL protein resulted in identification of small molecule inhibitors of ATPase activity. Binding assays using truncated SMARCA2 fragments confirmed novel allosteric binding site.

Figure 3b. Biochemical and biophysical investigation guided rational optimization resulted in identification of RVU-3626 used to build a hybrid PROTAC by linking to a ligand for the E3 ligase.

RYVU PROTAC LEADS TO SELECTIVE SMARCA2 DEGRADATION IN PROTEASOME DEPENDENT MANNER

a) Characterisation of RVU PROTAC

RVU	5363
hSMARCA2 pIC50	6.20
hSMARCA4 pIC50	6.20
SMARCA2 pD 1uM	10%
SMARCA2 pD 1uM	46%
SMARCA2 Ternary Complex Formation EC50 [uM]	0.15
MST Kd [uM]	0.70

b) Dose dependent SMARCA2/4 degradation

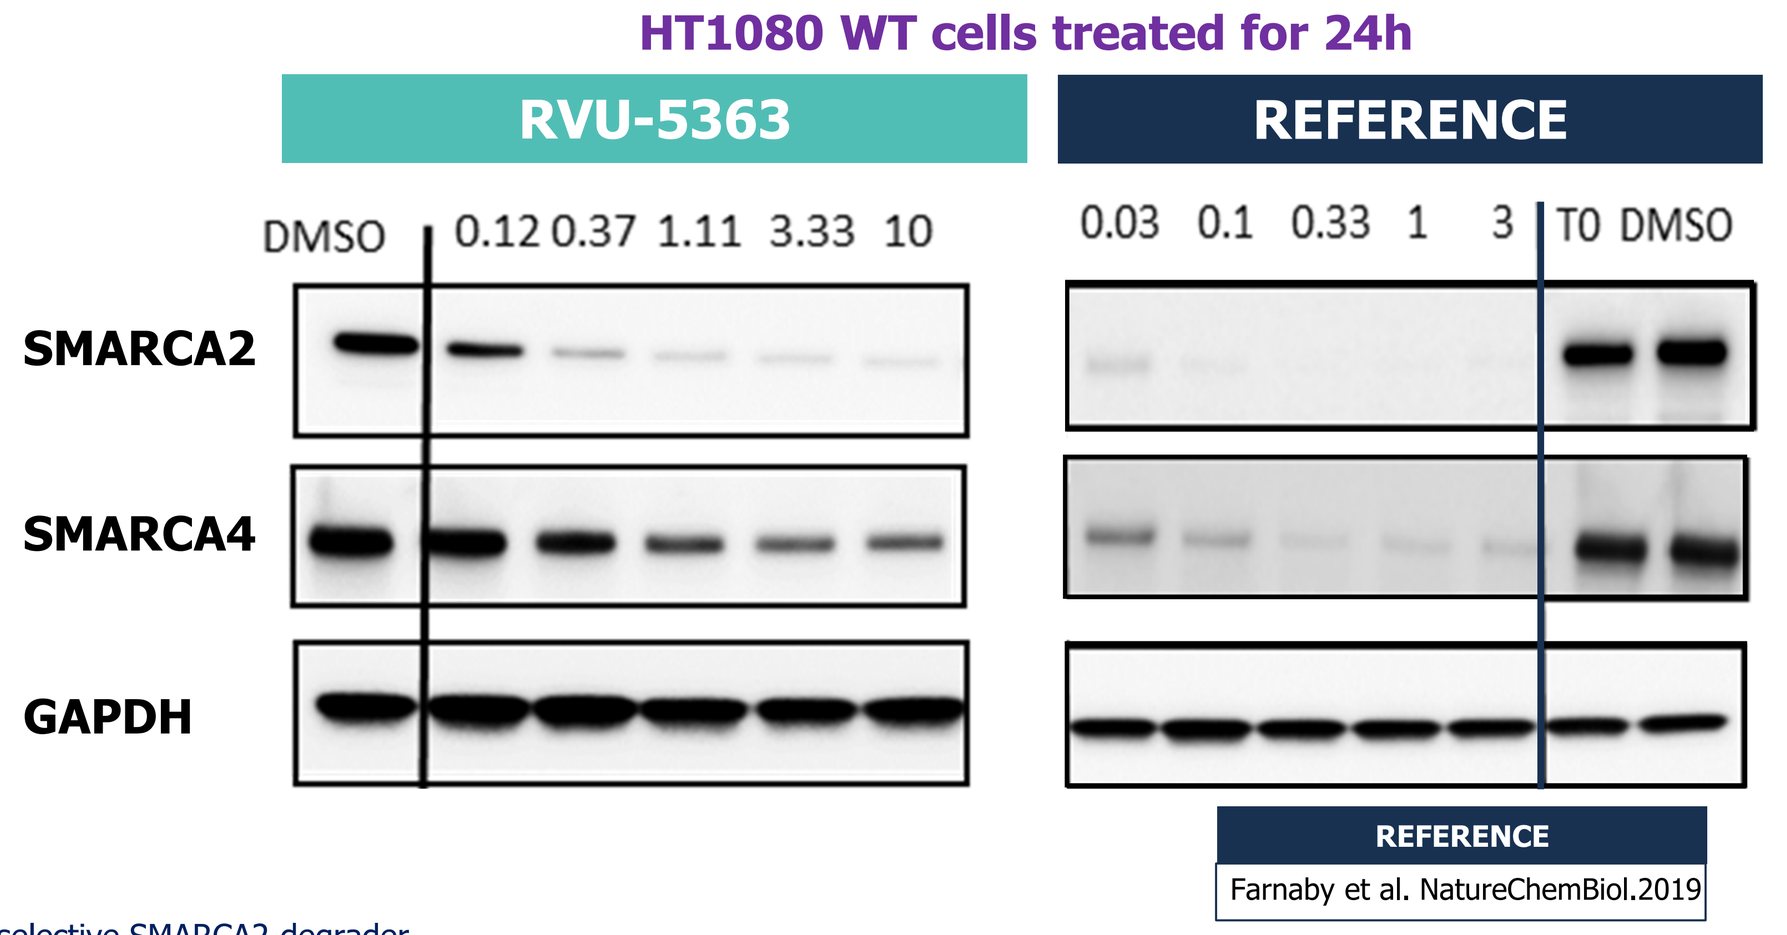

HT1080 WT cells treated for 24h
RVU-5363
REFERENCE
DMSO
0.12 0.37 1.11 3.33 10
0.03 0.1 0.33 1 3
T0 DMSO
SMARCA2
SMARCA4
GAPDH
REFERENCE
Farnaby et al. NatureChemBiol.2019

c) Proteosomal dependent SMARCA2 degradation

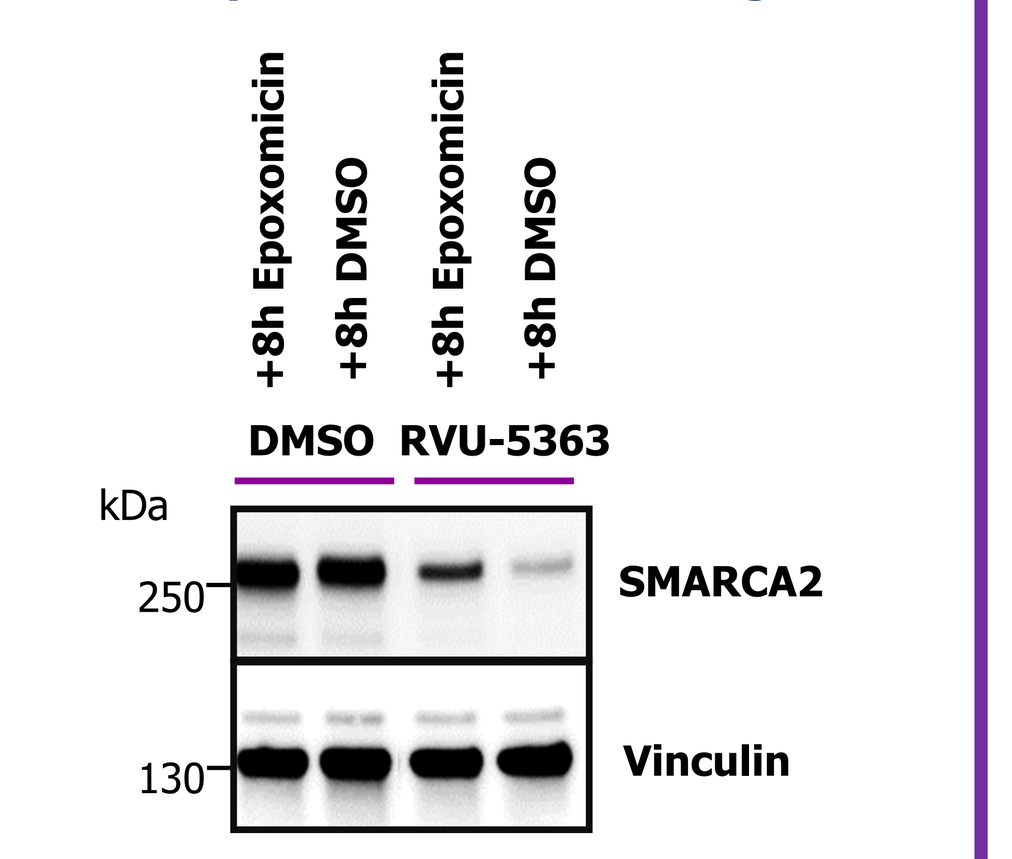

+8h Epoxomicin
+8h DMSO
+8h Epoxomicin
+8h DMSO
DMSO
RVU-5363
kDa
250
130
SMARCA2
Vinculin

Figure 4a. RVU-5363 is an equipotent SMARCA2/4 inhibitor and selective SMARCA2 degrader.
Figure 4b. Western blot analysis of HT1080 WT cells treated for 24h with indicated compounds (representative blots are shown).
Figure 4c. Western Blot analysis of HT1080 WT cells pretreated for 24h with DMSO or RVU-5363 followed by 8h incubation with DMSO or Epoxomicin (representative blots are shown).

BIOMARKER MODULATION CONFIRMS ON-TARGET ACTIVITY OF RYVU SERIES

a) RNAseq analysis in HT1080 isogenic cells

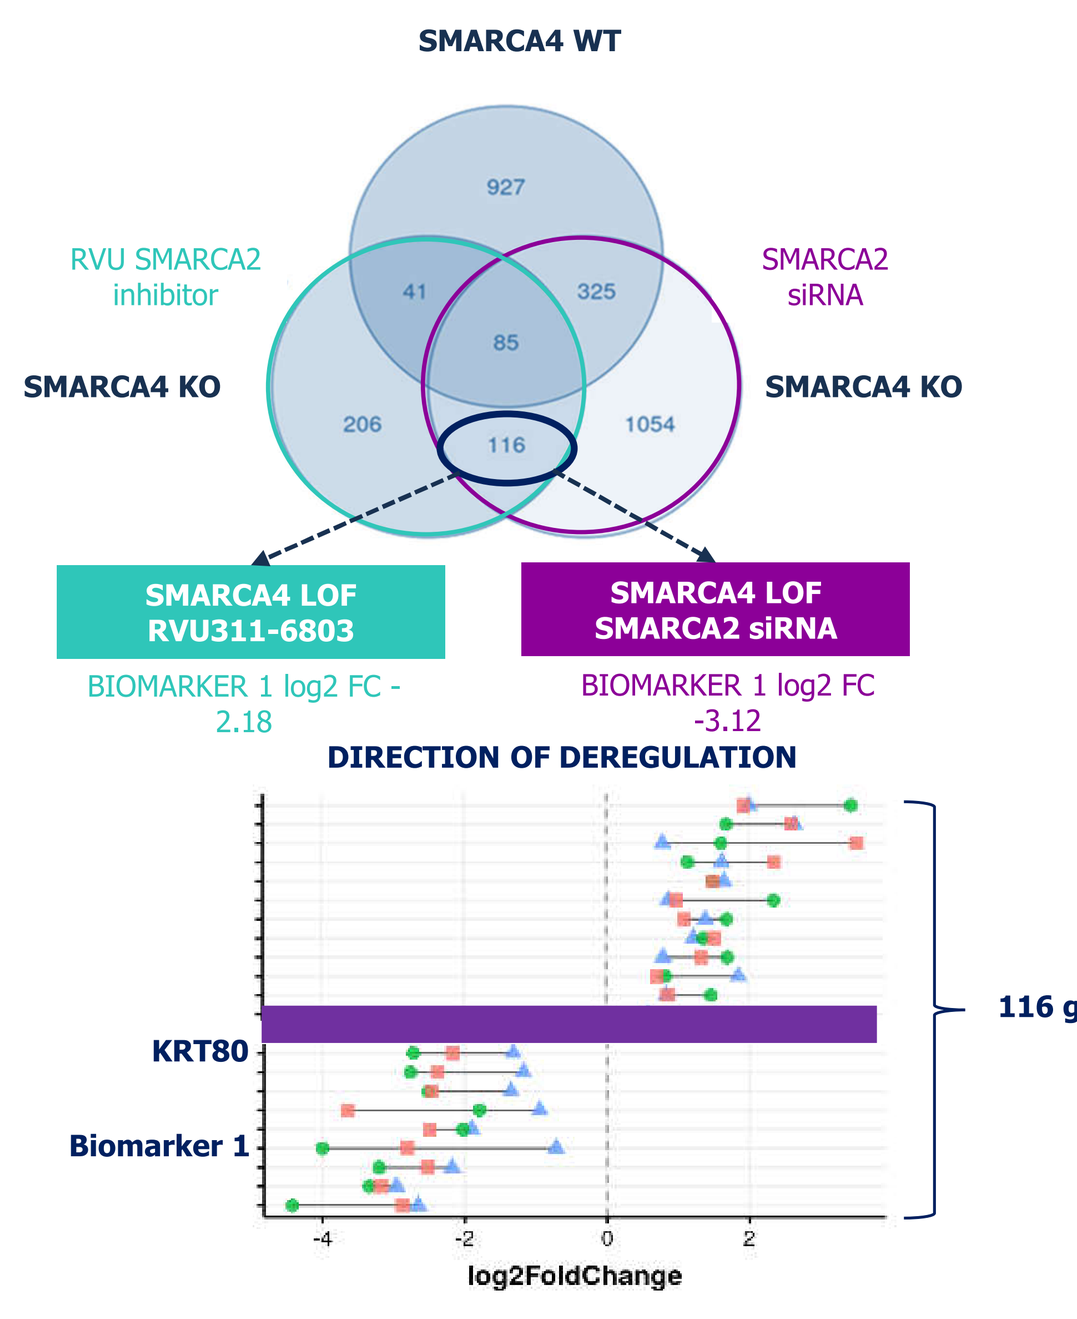

SMARCA4 WT
RVU SMARCA2 inhibitor
SMARCA2 siRNA
SMARCA4 KO
SMARCA4 KO
927
41
325
85
206
116
1054
SMARCA4 LOF RVU311-6803
SMARCA4 LOF SMARCA2 siRNA
BIOMARKER 1 log2 FC -2.18
BIOMARKER 1 log2 FC -3.12
DIRECTION OF DEREGULATION
116 genes
KRT80
Biomarker 1
log2FoldChange

b) Ryvu's PROTACs decrease KRT80 and Biomarker 1 in a dose dependent manner

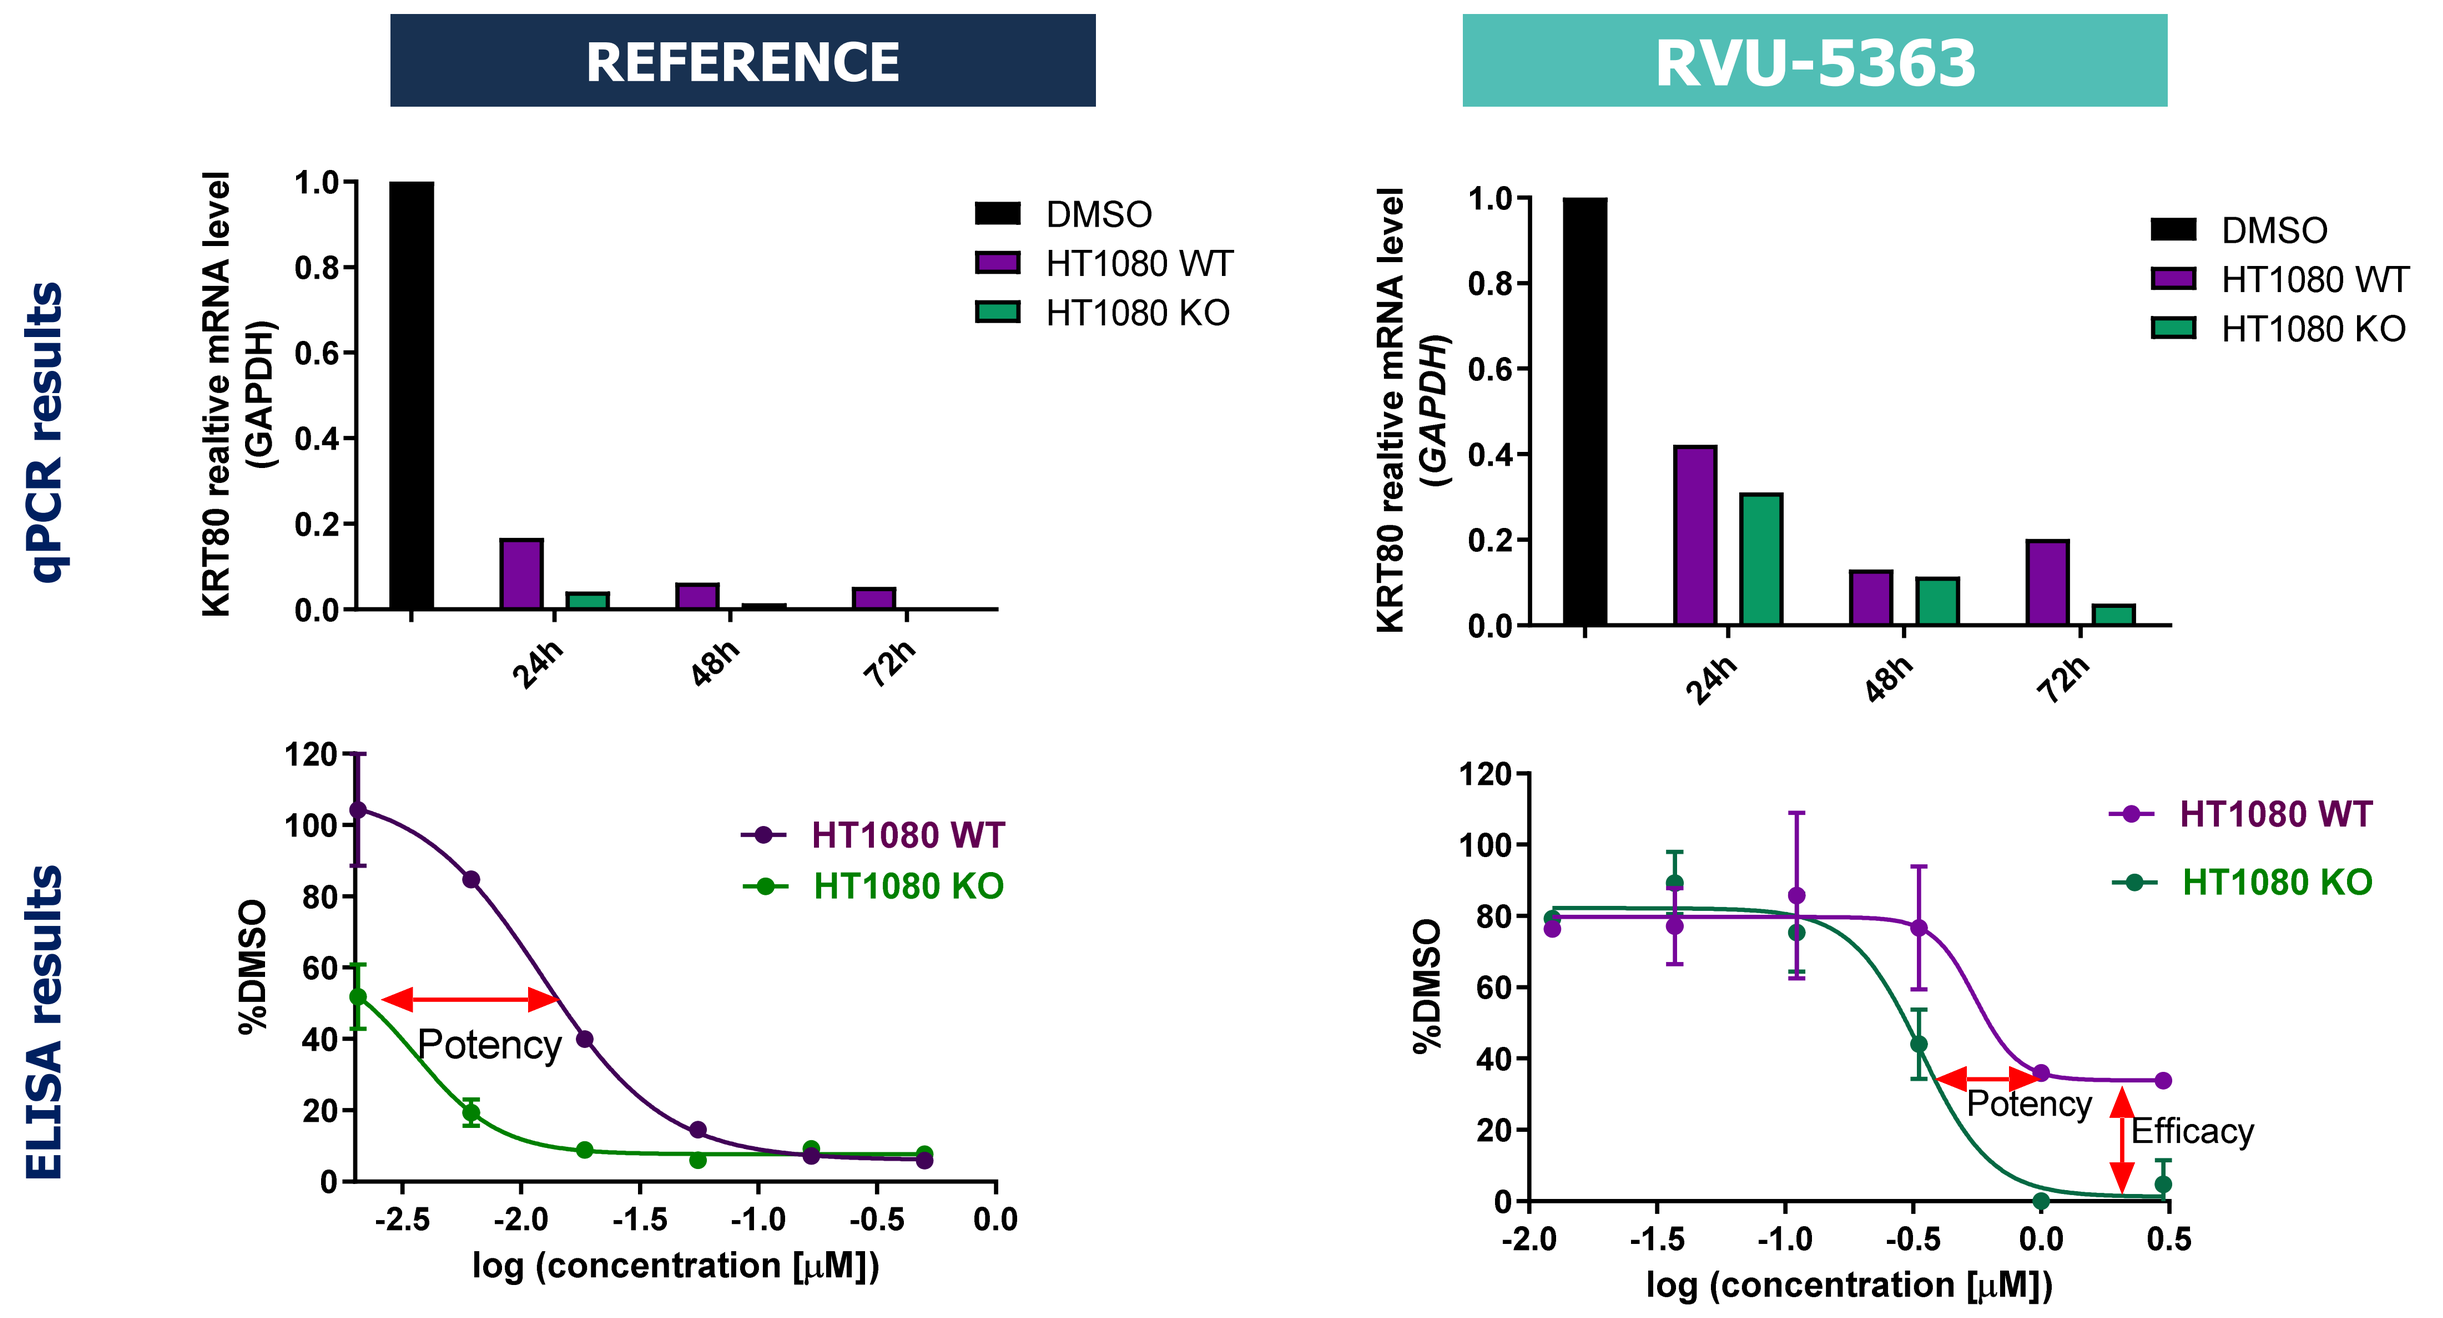

REFERENCE
RVU-5363
qPCR results
ELISA results
KRT80 realtive mRNA level (GAPDH)
DMSO
HT1080 WT
HT1080 KO
24h
48h
72h
%DMSO
Potency
Efficacy
log (concentration [µM])

Figure 5a. RNA sequecing in SMARCA4 WT and KO cells treated with SMARCA2 siRNA or small molecule SMARCA inhibitors allowed identification of 116 differentially expressed genes including KRT80 and Biomarker 1 (undisclosed).

Figure 5b. Down-regulation of KRT80 expression and Biomarker 1 expression post treatment with RVU-5363 or reference compound (values are expressed as means +/-SD).

RYVU PROTAC INDUCES APOPTOSIS PEREDOMINANTLY IN SMARCA4 KO CELLS

a) Long-lasting selective SMARCA2 degradation

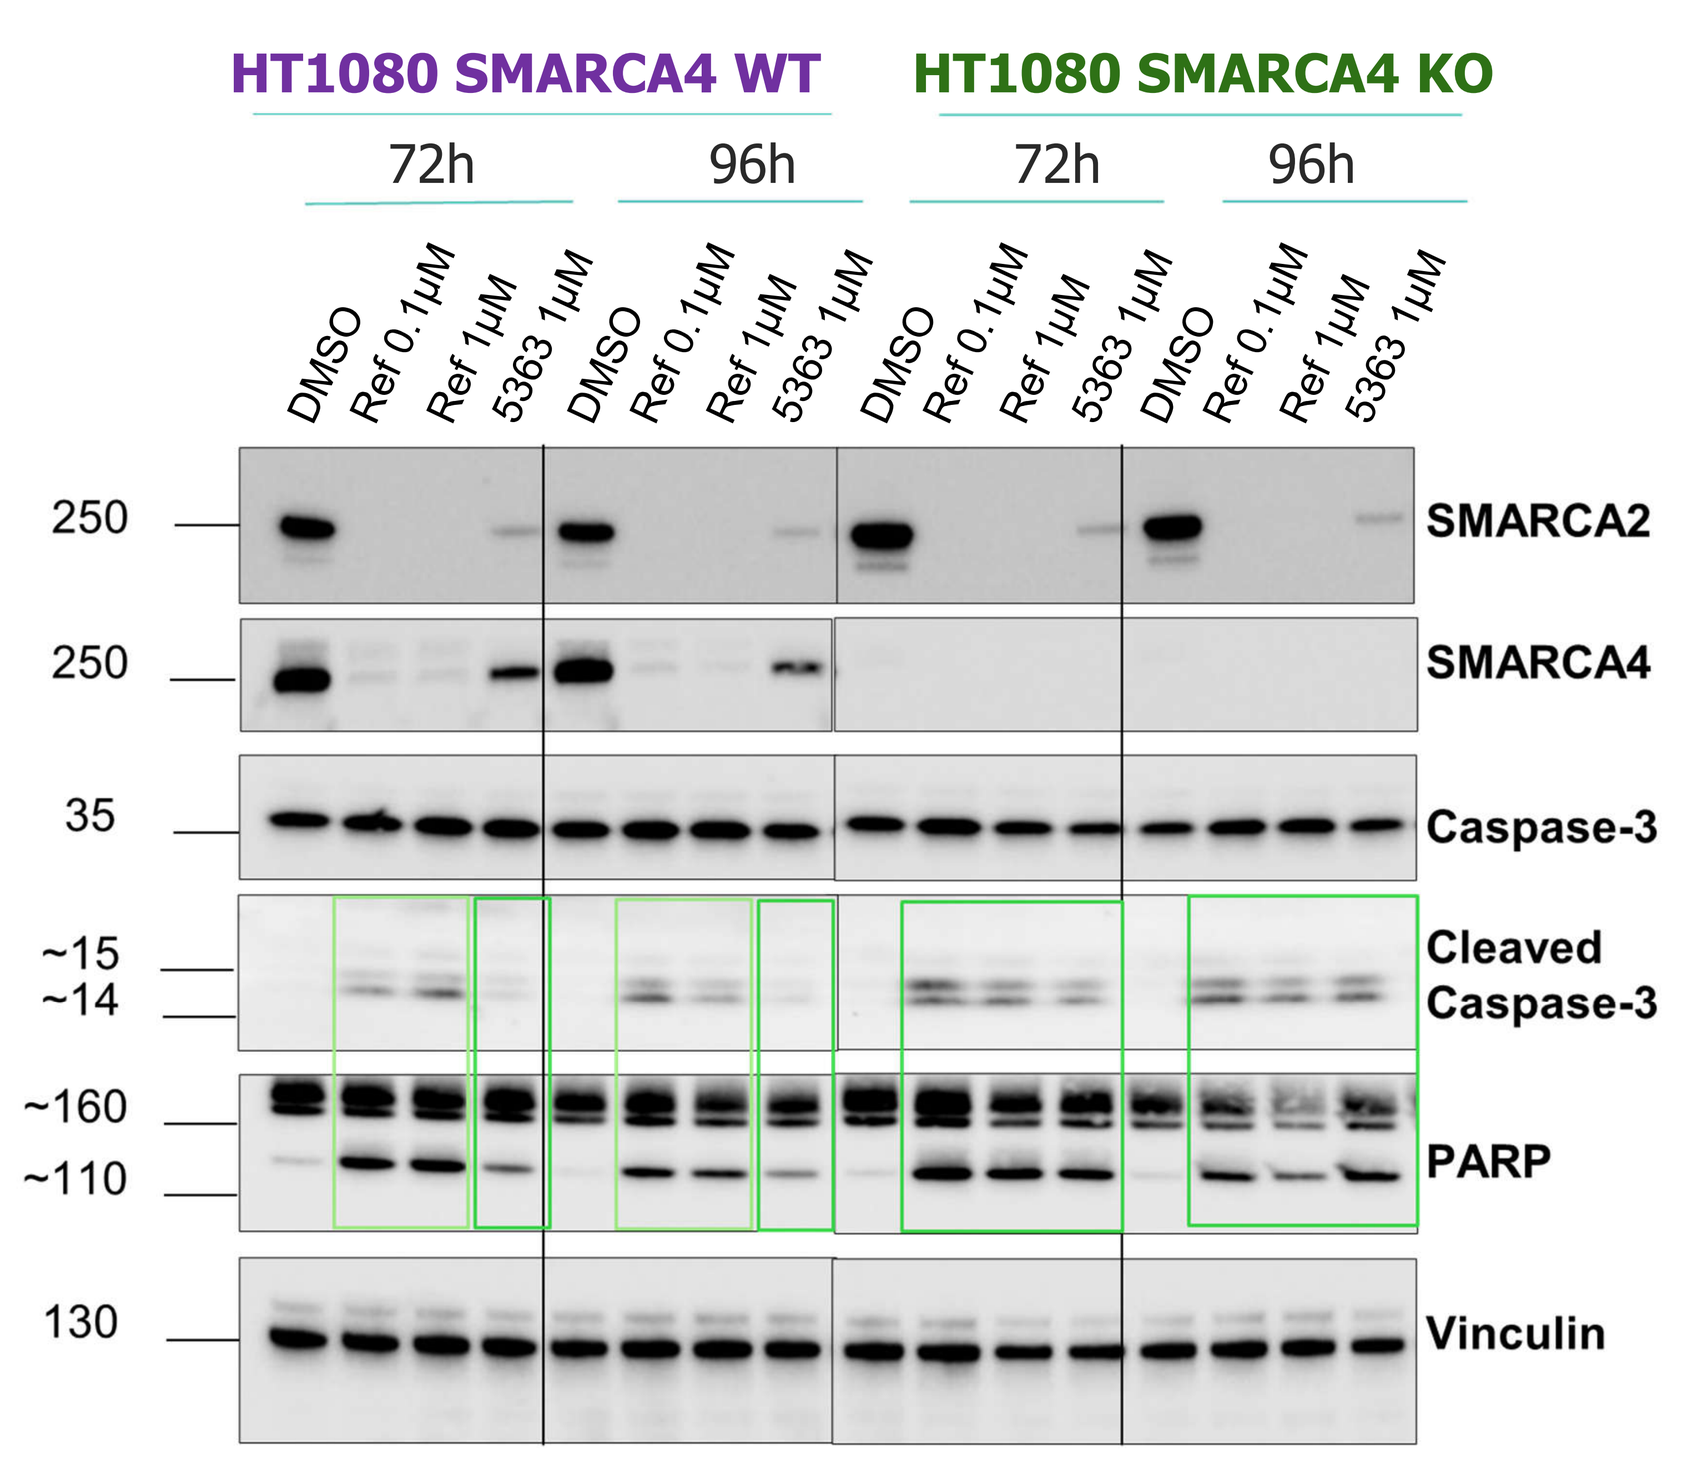

HT1080 SMARCA4 WT
HT1080 SMARCA4 KO
72h
96h
DMSO
Ref 0.1µM
Ref 1µM
5363 1µM
SMARCA2
SMARCA4
Caspase-3
Cleaved Caspase-3
PARP
Vinculin

b) Induction of apoptotic pathway in SMARCA4 knock-out cells by RVU-5363

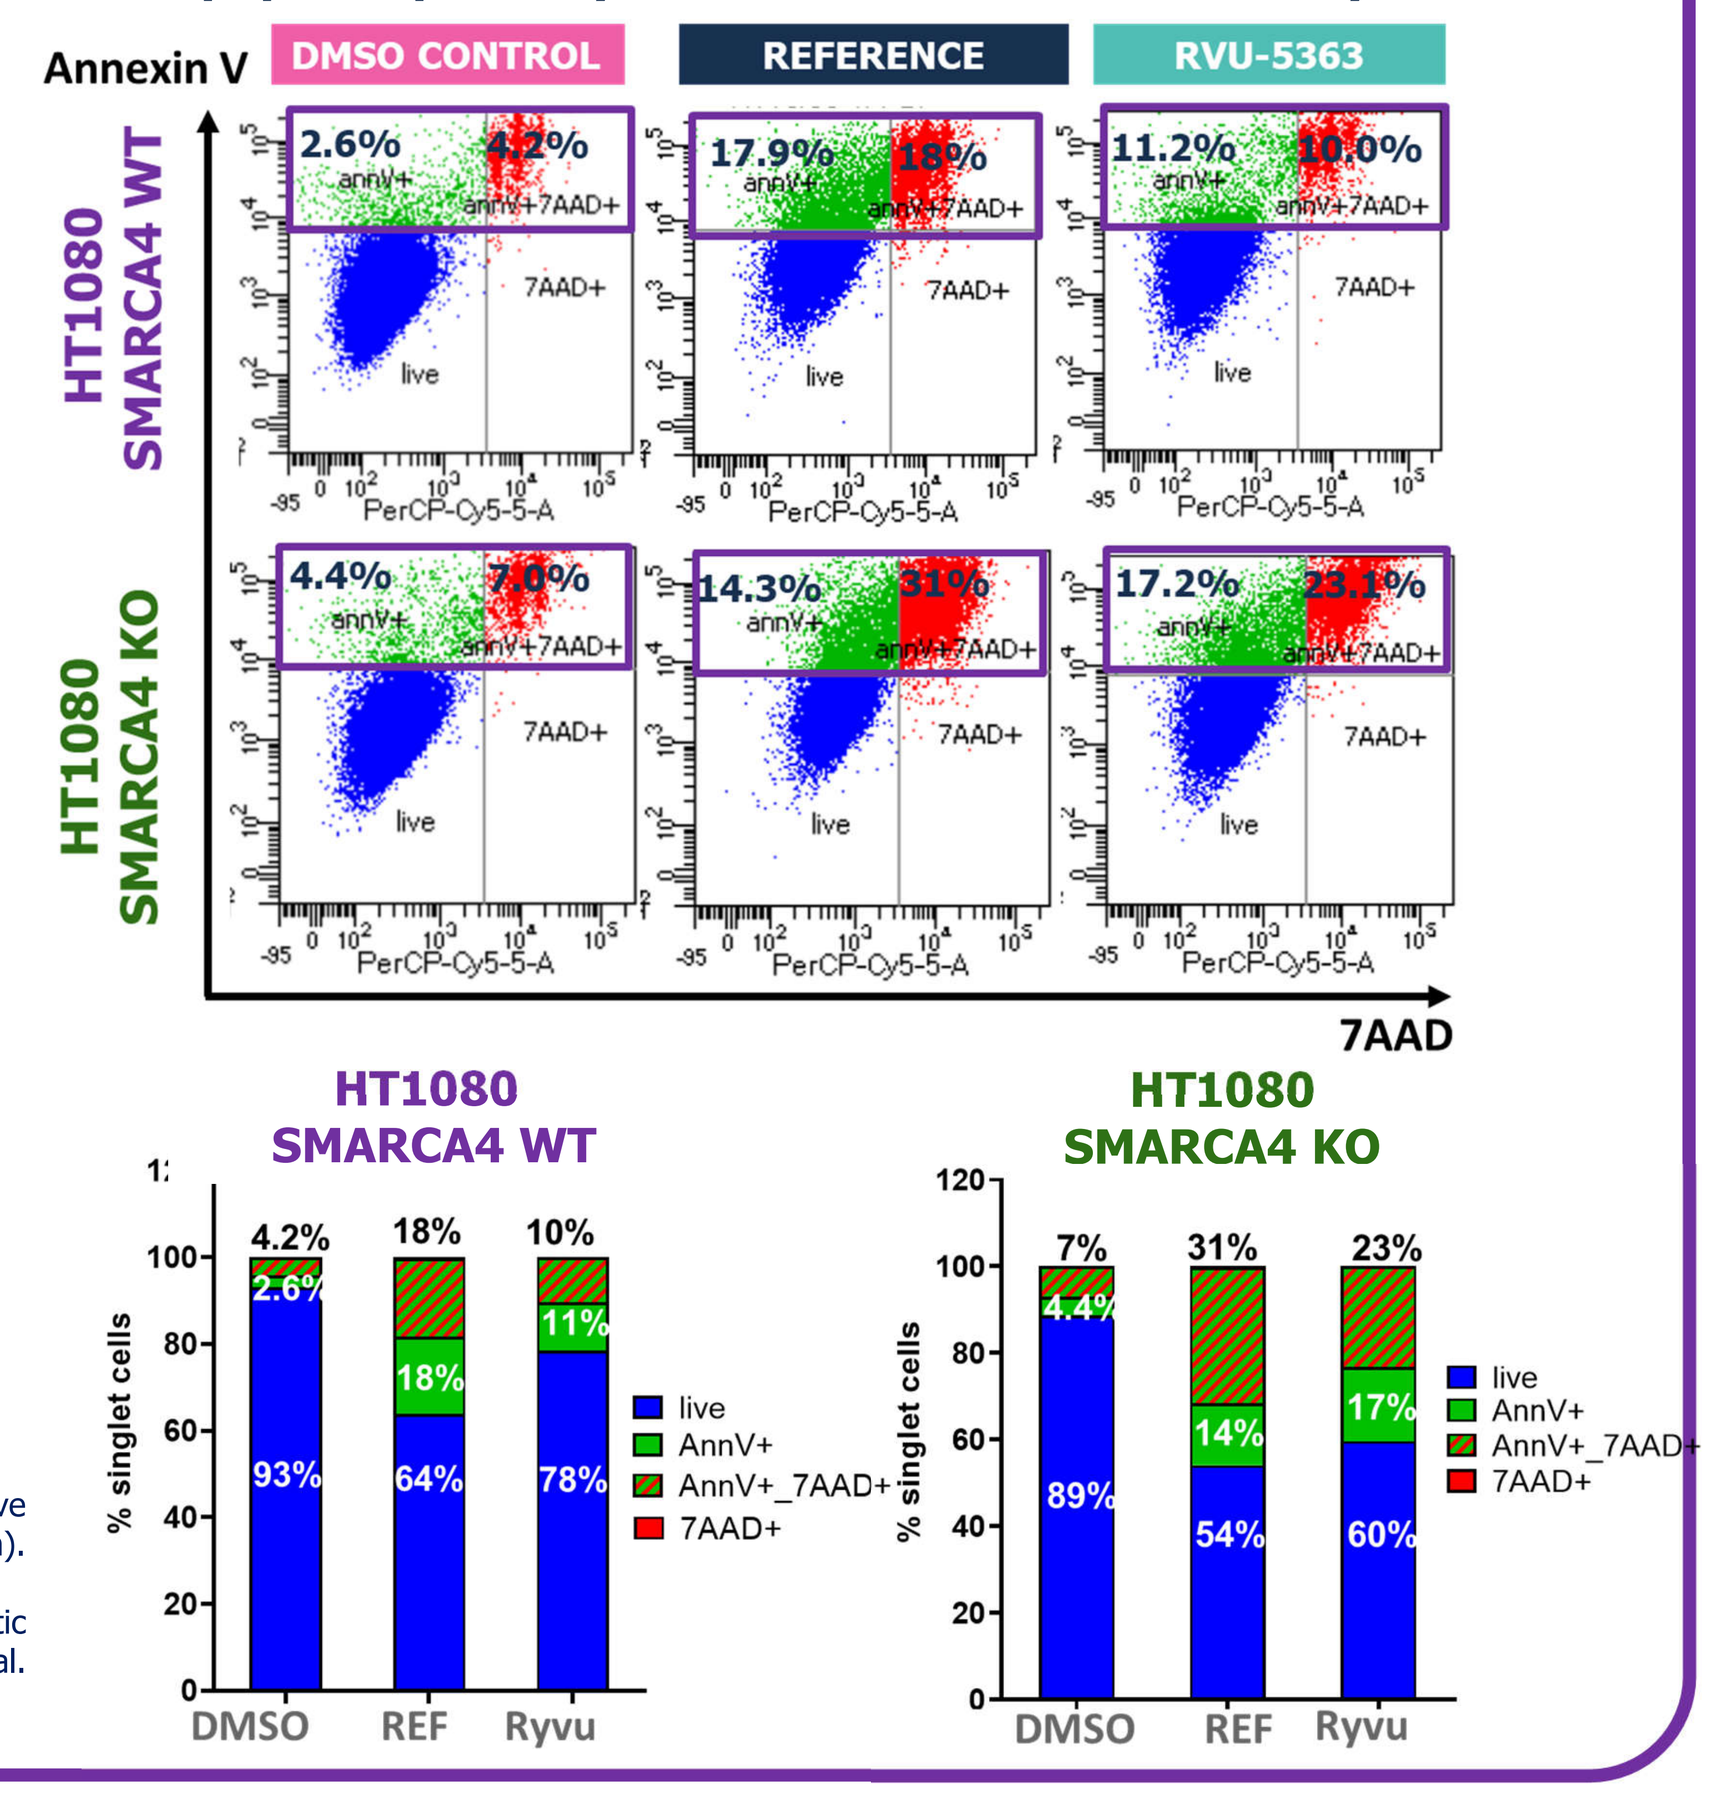

Annexin V
DMSO CONTROL
REFERENCE
RVU-5363
HT1080 SMARCA4 WT
HT1080 SMARCA4 KO
7AAD
HT1080 SMARCA4 WT
HT1080 SMARCA4 KO
% singlet cells
live
AnnV+
AnnV+_7AAD+
7AAD+
DMSO
REF
Ryvu

Figure 6a. Western blot analysis of HT1080 isogenic cells at 72h and 96h post treatment with indicated compounds showing long lasting and selective SMARCA2 degradation for Ryvu PROTAC, inducing Caspase and PARP cleavage predominantly in SMARCA4 KO cells (representative blots are shown).

Figure 6b. FACS analysis at 72h post treatment with indicated compounds showing induction of early apoptotic (AnnexinV+ve) and late apoptotic cells (AnnexinV+ve7AAD+ve predominantly in SMARCA4 KO cells in comparison to non-selective reference compound (Farnaby et al. NatureChemBiol.2019).

RYVU SMARCA2 PROTAC SELECTIVELY KILLS SMARCA4 KO CELLS

2D ASSAY

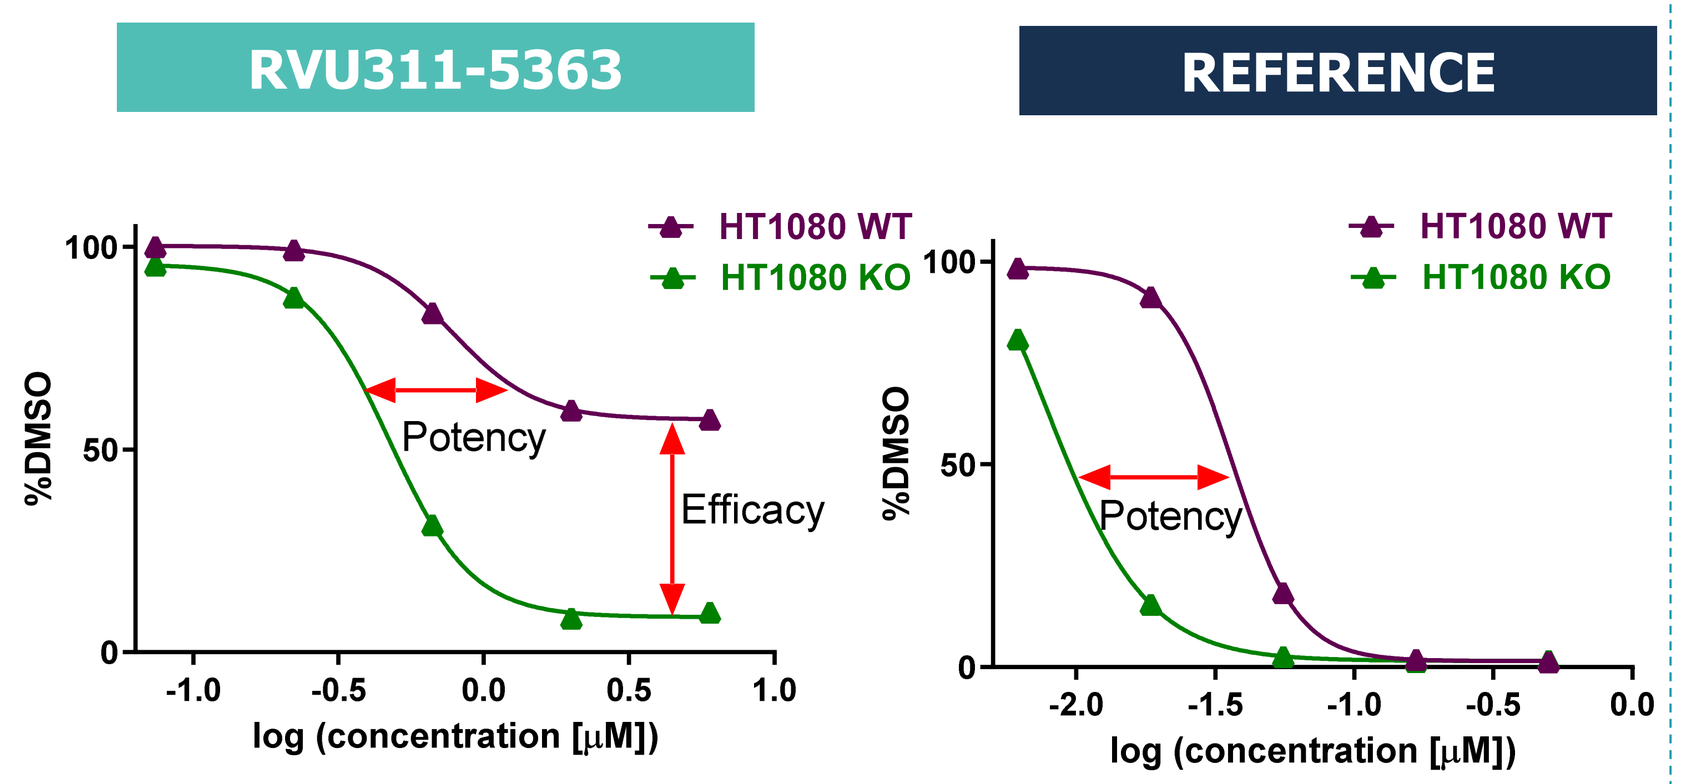

RVU311-5363
REFERENCE
HT1080 WT
HT1080 KO
%DMSO
Potency
Efficacy
log (concentration [µM])

3D ASSAY

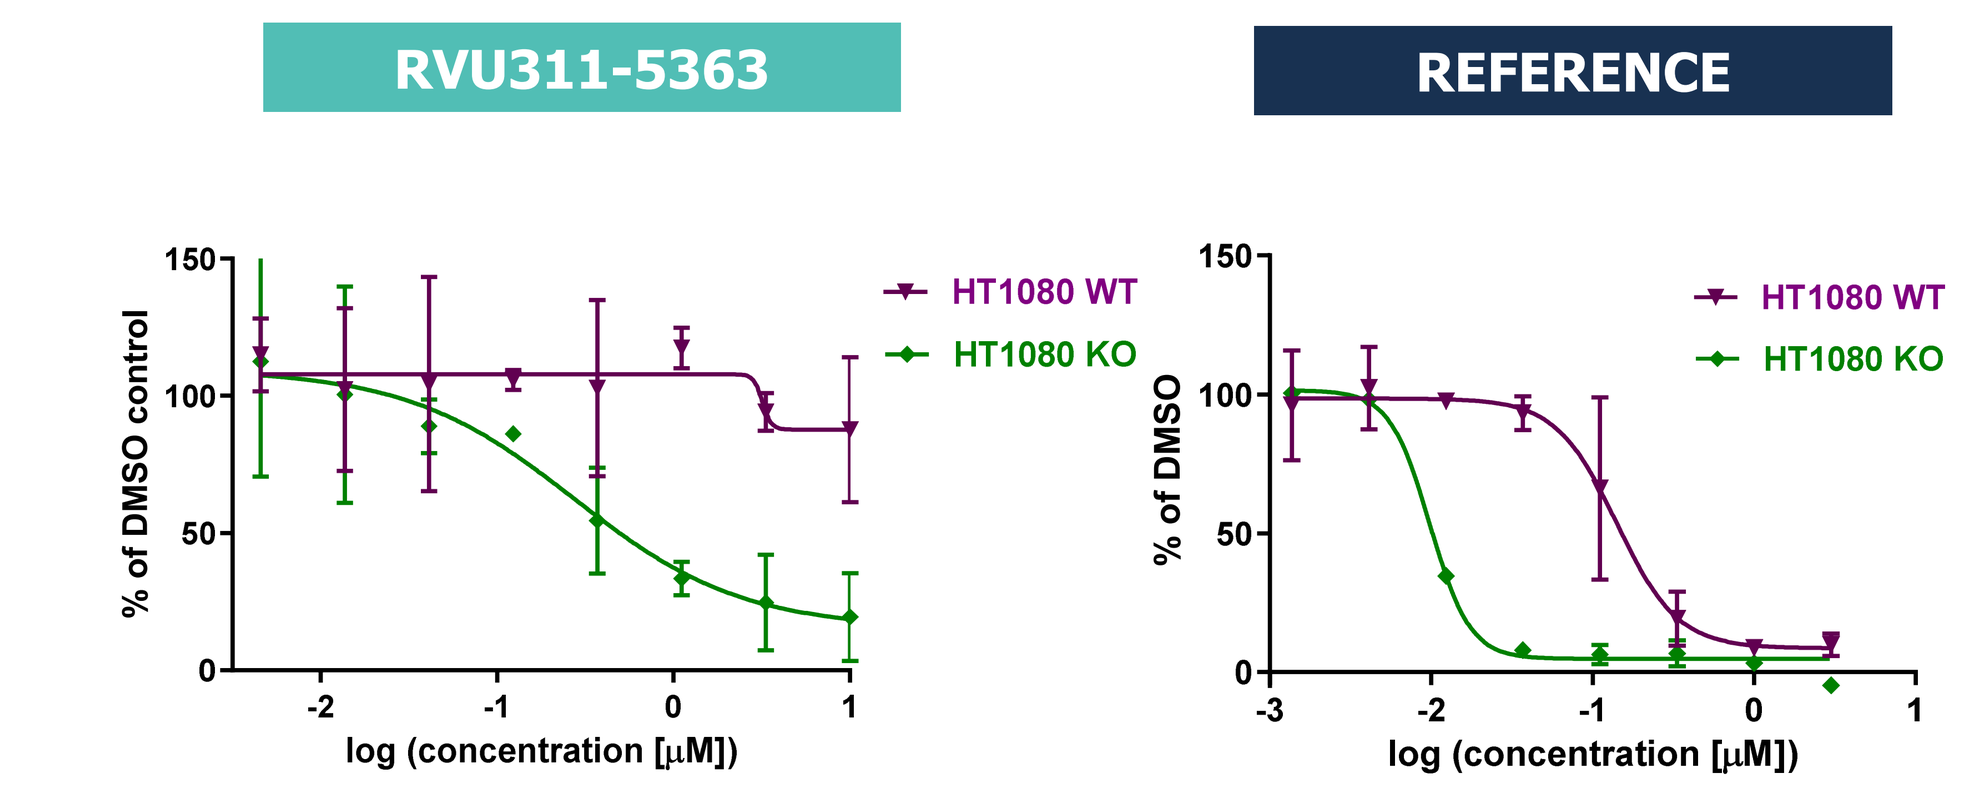

RVU311-5363
REFERENCE
HT1080 WT
HT1080 KO
% of DMSO control
log (concentration [µM])

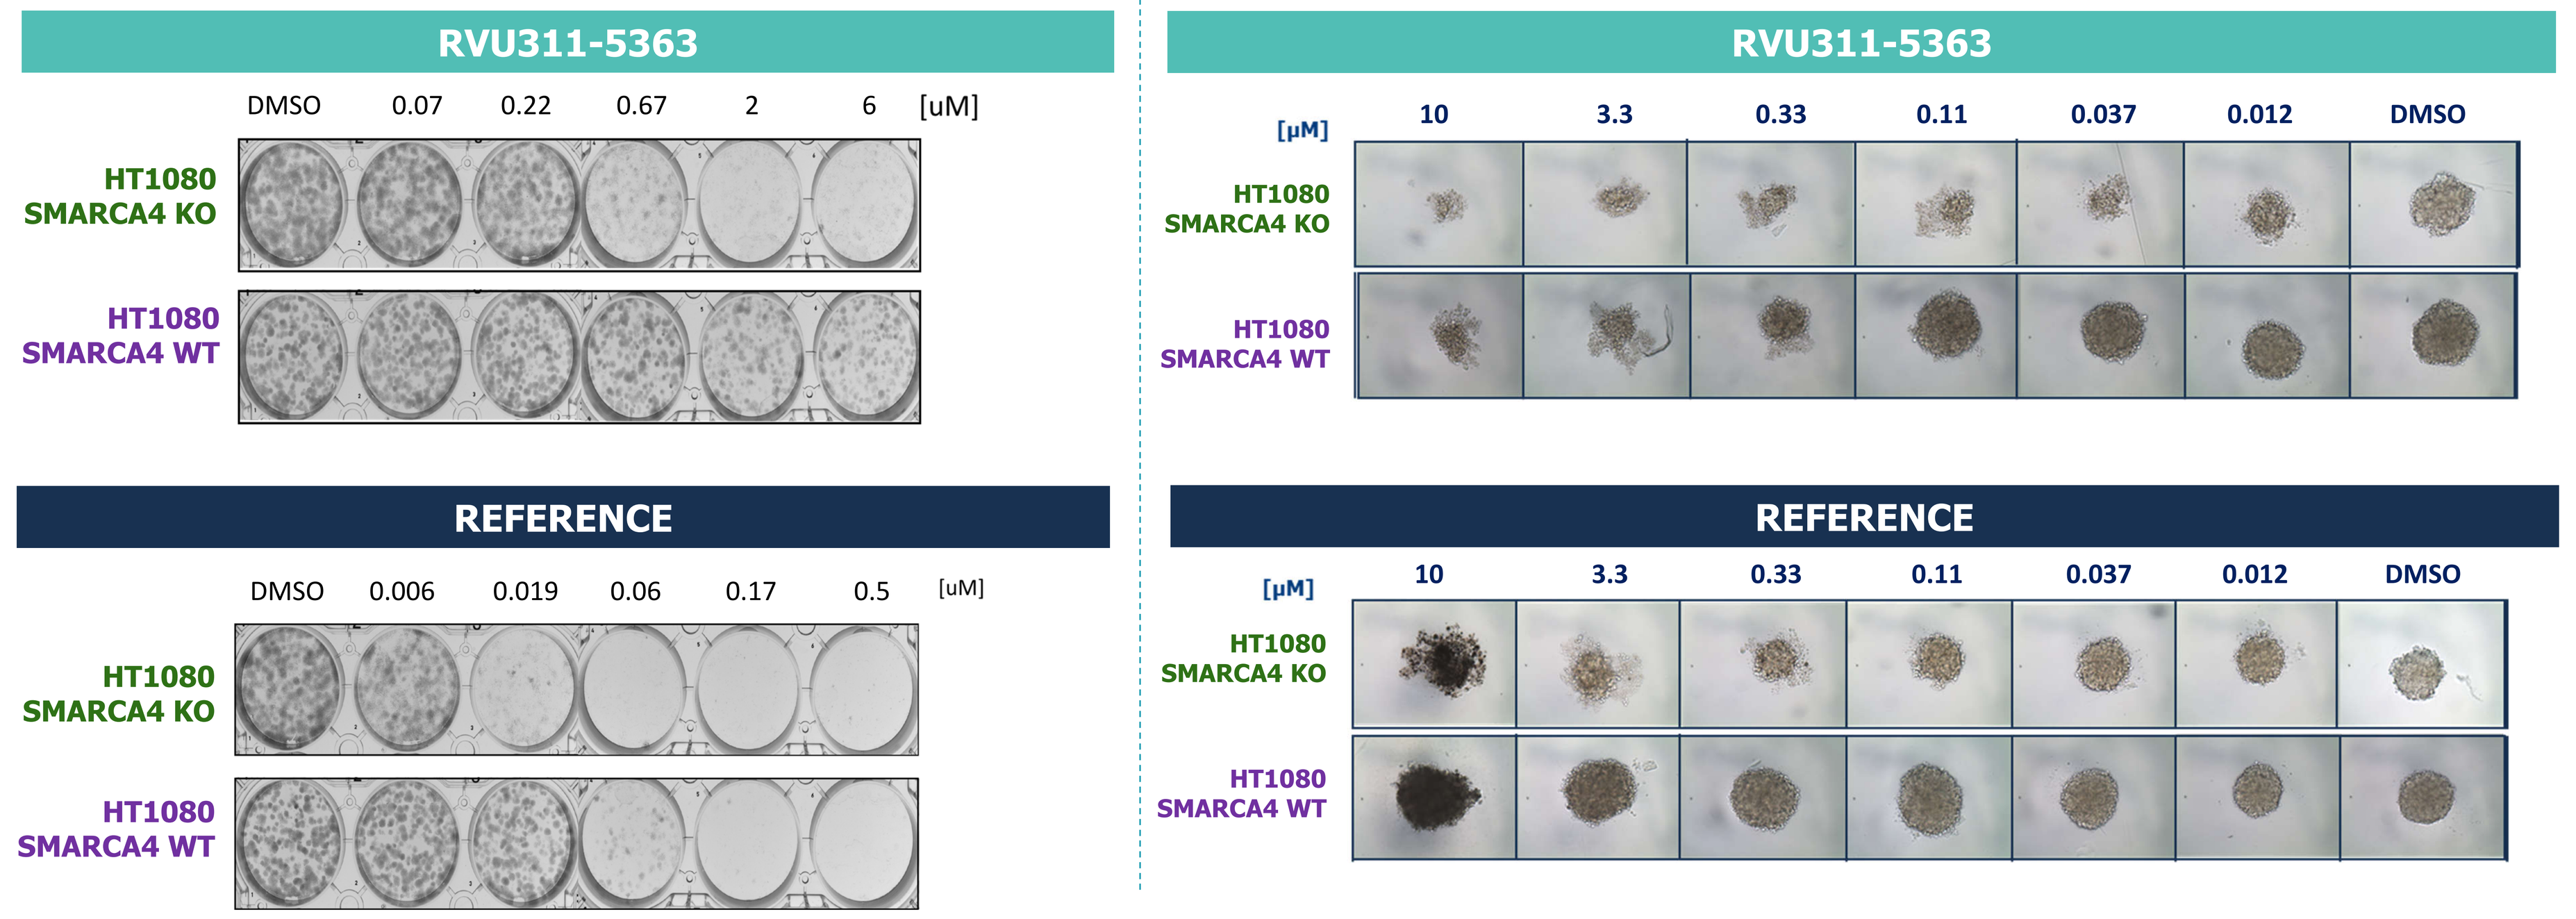

RVU311-5363
DMSO 0.07 0.22 0.67 2 6 [uM]
HT1080 SMARCA4 KO
HT1080 SMARCA4 WT
REFERENCE
DMSO 0.006 0.019 0.06 0.17 0.5 [uM]
RVU311-5363
[µM] 10 3.3 0.33 0.11 0.037 0.012 DMSO
REFERENCE

Isogenic HT1080 pair

				RVU311-5363	Fold difference	Reference	Fold difference
2D	HT1080 WT	SMARCA2 WT	SMARCA4 WT	>6 µM	>13x	0.036 µM	5x
	HT1080 KO	SMARCA2 WT	SMARCA4 KO	0.47 µM		0.007 µM	
3D	HT1080 WT	SMARCA2 WT	SMARCA4 WT	>6 µM	>22x	0.14 µM	14x
	HT1080 KO	SMARCA2 WT	SMARCA4 KO	0.27 µM		0.01 µM	

Figure 7 Left panel. HT1080 isogenic cells were treated for 7 days with indicated compounds. At day 7 Alamar Blue assay was performed (top panel) and colonies were stained with Crystal violet (values are expressed as means +/-SD).

Figure 7 Righ panel. HT1080 isogenic cells were seeded form 3D spheroids in the presence of indicated compounds. At day 7 images were taken and viability was assesed usng CellTiter-Glo@3D (values are expressed as means +/-SD).

CONCLUSIONS

- **We identified first in class allosteric inhibitors of SMARCA2/4 ATPase activity.**
- **Rational guided optimization resulted in identification of nM inhibitor used to built a hybrid PROTAC by linking to a ligand for the E3 ligase.**
- **Treatment with our PROTAC led to selective and long-lasting, proteasomal dependent degradation of SMARCA2 protein.**
- **We identified selective SMARCA2 biomarkers and confirmed their modulation with full efficacy in SMARCA4 KO cells.**
- **Selective SMARCA2 degradation led to induction of apoptotic pathway and in consequence to a targeted cell death of SMARCA4 mutated cancers.**
- **Fine-tuning of available PROTACs will allow for proof-of-concept experiments in animal models as a single agent or in combinations with radio- or chemotherapies.**
- **Taken together, these data provides rationale for a novel targeted therapy that may lead to durable responses in NSCLC patients carrying SMARCA4 LOF mutations.**

Republic of Poland · The National Centre for Research and Development · Project supported by the European Regional Development Funds under the Measure 1.1.1. Operational Program - Smart Growth. (POIR.01.01.01-00-0647/19-00) · European Funds Smart Growth · European Union